DINAMO – DIabetes study of liNAgliptin and eMpagliflozin in children and adOlescents with Type 2 Diabetes (T2D): Innovative Study Design and Baseline Characteristics

Lori Laffel,[1] Thomas Danne,[2] William Tamborlane,[3] Georgeanna Klingensmith,[4] Christy Schroeder,[5] Dietmar Neubacher,[6] Nima Soleymanlou,[5] Jan Marquard,[5] Philip Zeitler,[7] Steven Willi[8]

[1]Joslin Diabetes Center, Harvard Medical School, Boston, MA, USA; [2]Auf der Bult Kinder-und Jugendkrankenhaus, Hannover, Germany; [3]Yale University, New Haven, CT, USA; [4]Barbara Davis Center, University of Colorado, Aurora, CO, USA; [5]Boehringer Ingelheim Pharmaceuticals Inc, Ridgefield, CT, USA; [6]Boehringer Ingelheim Pharma GmbH & Co KG, Biberach, Germany; [7]University of Colorado Anschutz Medical Center, Aurora, CO, USA; [8]Children's Hospital of Philadelphia, Philadelphia, PA, USA

BACKGROUND

- Over the past 25 years, more than 4 classes of new drugs for diabetes treatment have been approved for adults with type 2 diabetes (T2D). In contrast, over the same period, only 1 new drug class, glucagon-like peptide-1 (GLP-1) agonists, has been approved for use in children and adolescents with T2D.[1,2]
- T2D in children and adolescents is challenging to manage, creating a pressing need for an expanded treatment armamentarium. The DINAMO study (NCT03429543) was designed to overcome the recruitment difficulties that plagued previous T2D studies in children and adolescents.
- Consequently, we present herein the novel study design and baseline clinical characteristics of children and adolescents with T2D enrolled in the pivotal trial of both linagliptin, a dipeptidyl peptidase-4 (DPP-4), and empagliflozin, a sodium-glucose co-transporter-2 (SGLT2) inhibitor.

Scan QR code for an **interactive** electronic device-friendly copy of the poster

OBJECTIVES

- The main objective of the DINAMO study is to assess the efficacy and safety of a single dose of linagliptin and an empagliflozin dosing regimen versus placebo after 26 weeks of treatment in children and adolescents with T2D managed with metformin and/or insulin or lifestyle modification alone.
- In addition, this study will assess the safety of linagliptin and empagliflozin over 52 weeks of treatment.

METHODS

- Multicenter, randomized, double-blind, placebo-controlled, and parallel group design of 3 treatment arms (placebo, linagliptin 5 mg, empagliflozin 10 mg) over 26 weeks.
- Dose increase of empagliflozin 10 mg to 25 mg at Week 14 in participants not achieving glycated hemoglobin (HbA1c) <7.0% at Week 12.
- Double-blind active treatment safety extension period up to 52 weeks (**Figure 1**).
- Participants on placebo re-randomized at Week 26 to receive either linagliptin or 1 of the empagliflozin doses (empagliflozin 10 mg or 25 mg).

Figure 1. Study design

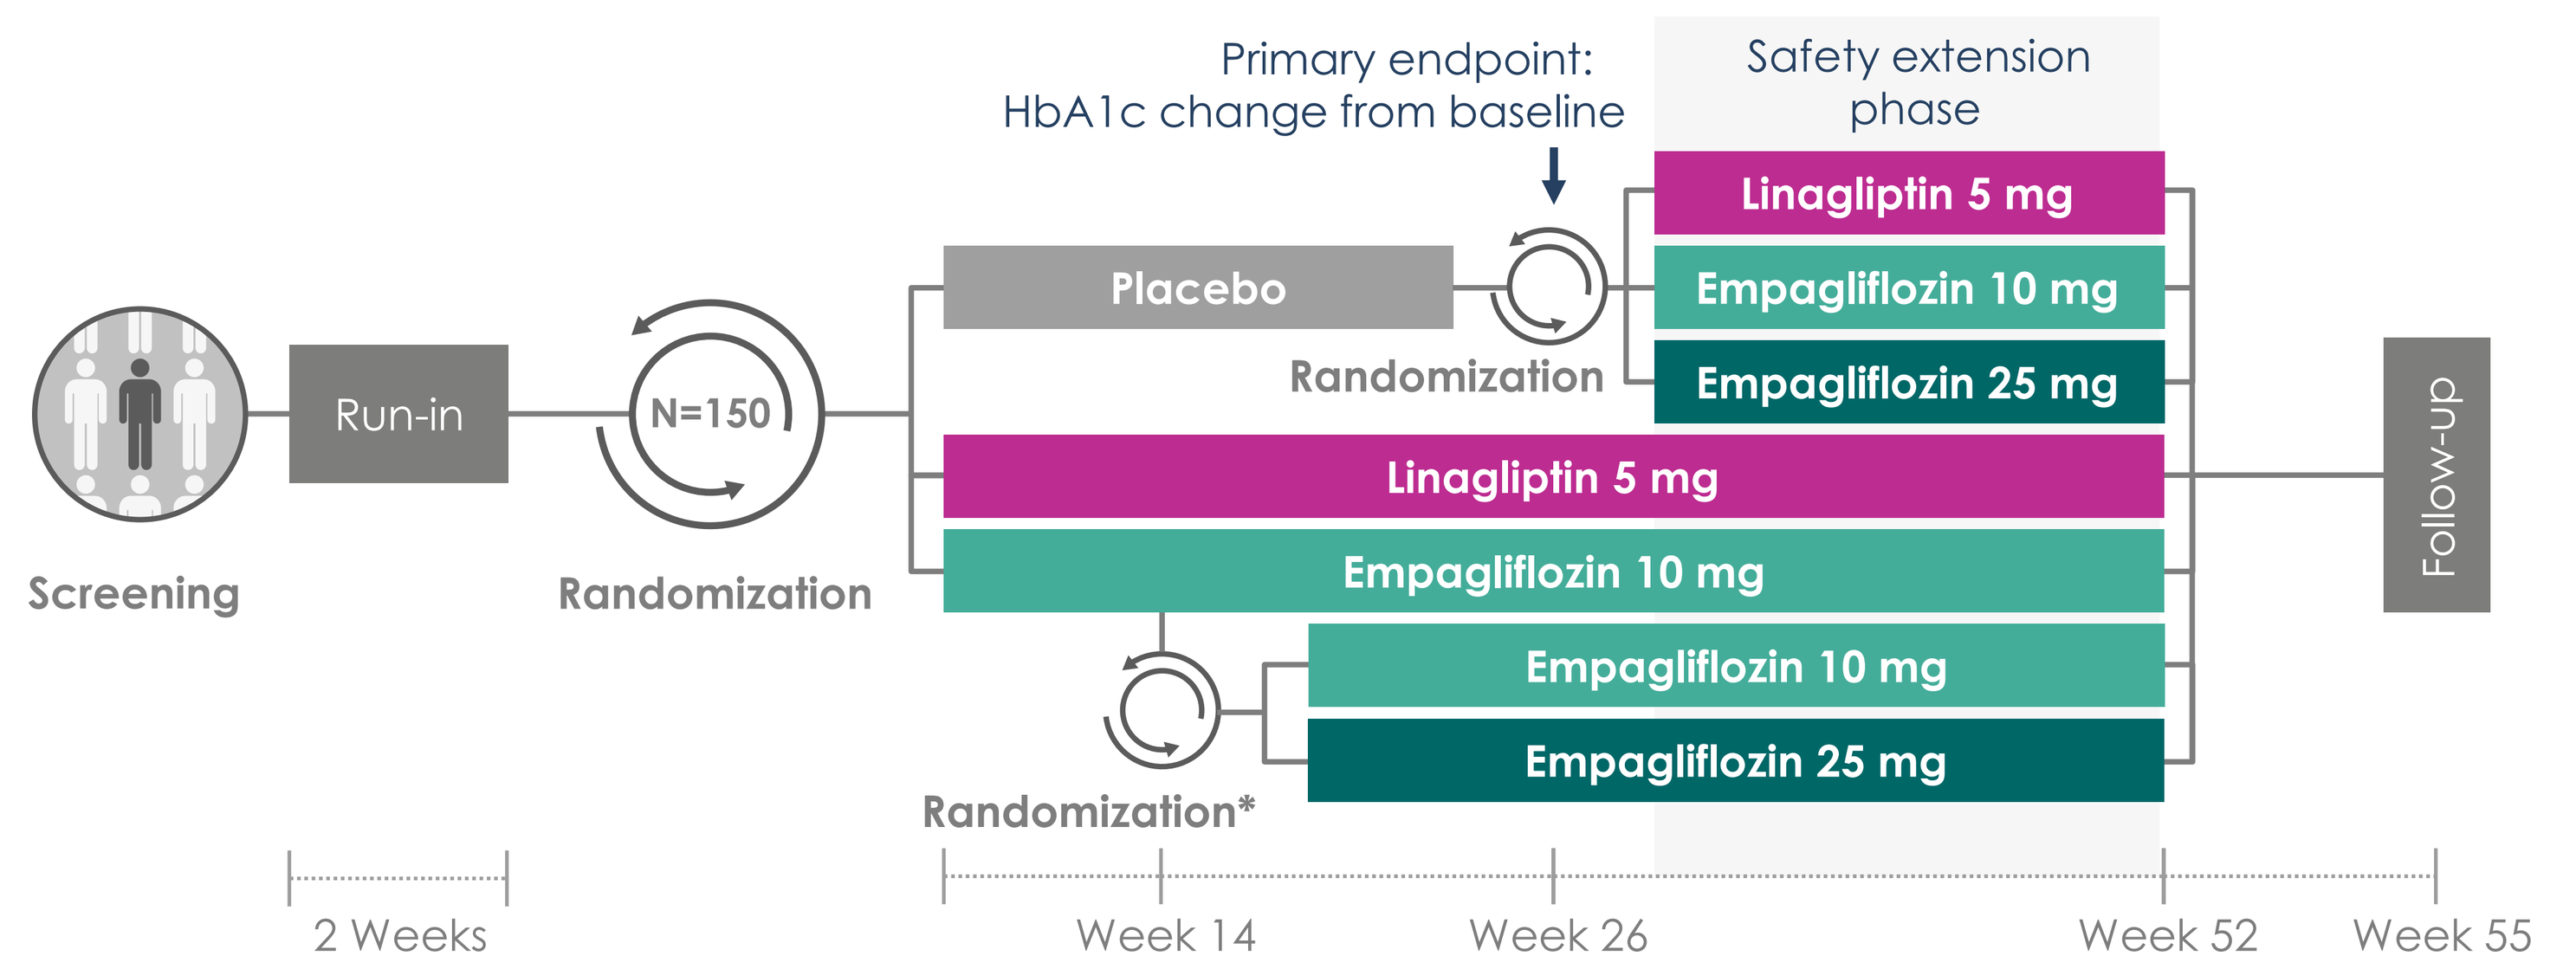
Primary endpoint: HbA1c change from baseline
Safety extension phase
Linagliptin 5 mg
Placebo
Empagliflozin 10 mg
Randomization
Empagliflozin 25 mg
Run-in
N=150
Linagliptin 5 mg
Empagliflozin 10 mg
Empagliflozin 10 mg
Empagliflozin 25 mg
Follow-up
Screening
Randomization
Randomization*
2 Weeks
Week 14
Week 26
Week 52
Week 55

*Re-randomization at Week 14 for patients not achieving HbA1c <7% at Week 12.
HbA1c, glycated hemoglobin.

- Since participants and investigators remain blinded, investigators will perform a re-randomization for all participants at Week 14 and Week 26 to get new trial medication kits assigned.
- The sample size estimate targets 150 participants randomized 1:1:1 to linagliptin 5 mg, empagliflozin 10 mg, and placebo.

- Potential participants who have a modifiable exclusion on initial screening (e.g., HbA1c <6.5%) can be rescreened up to 5 times.
- A dedicated Study Steering Committee is involved in design, development, and finalization of the protocol, ongoing review of study progress, and adjustment of the recruitment strategy as needed.
- An independent Data Monitoring Committee is responsible for reviewing unblinded safety and efficacy data.
- In the US, the study was conducted in collaboration with the U.S. Pediatric Diabetes Consortium (PDC), a network of approximately 30 US diabetes centers.

Main inclusion criteria

- Children and adolescents from 10 to 17 years of age (inclusive) at the time of randomization.
- Documented diagnosis of T2D for at least 8 weeks prior to screening.
- HbA1c ≥6.5% and ≤10.5% at screening.
- Patients treated with diet and exercise plus metformin at least 1000 mg/day (or up to a maximal tolerated dose) at a stable dose for 8 weeks prior to randomization and/or stable insulin therapy (basal or Multiple Daily Injection) for 8 weeks prior to randomization (stable insulin therapy is defined as a weekly average variation of the basal insulin dose ≤0.1 U/kg over 8 weeks prior to randomization).
- Patients not tolerating metformin (where insulin monotherapy would be the only alternative approved option) and treated with diet and exercise only.
- Body mass index (BMI) ≥85th percentile for age and sex according to World Health Organization references at run-in.
- Negative for both islet cell antigen auto-antibodies and glutamic acid decarboxylase as measured by the central laboratory at screening.
- Non-fasting serum C-peptide levels ≥0.6 ng/ml or >0.199 nmol/l as measured by the central laboratory at screening.

Main exclusion criteria

- History of acute metabolic decompensation such as diabetic ketoacidosis within 8 weeks prior to screening and up to randomization.
- Diagnosis of monogenic diabetes (e.g., maturity onset diabetes of the young [MODY]).
- Impaired renal function defined as estimated glomerular filtration rate <60 ml/min/1.73m² (according to Zappitelli formula) at screening.

Outcome measures

- The primary endpoint is the change in HbA1c (%) from baseline to the end of 26 weeks.
- Secondary endpoints from baseline to end of 26 weeks are:
 - Change in fasting plasma glucose (mg/dl)
 - Change in body weight (kg)
 - Change in systolic blood pressure (mmHg)
 - Change in diastolic blood pressure (mmHg)
 - Proportion of patients who achieve HbA1c <6.5% at the end of 26 weeks
 - Proportion of patients who achieve HbA1c <7.0% at the end of 26 weeks.

RESULTS

- Within 3 years, which included recruitment during the COVID-19 pandemic, DINAMO screened 263 participants and enrolled 158 participants aged 10 to <18 years in 12 countries (**Figures 2** and **3**). The 158 participants were randomized at 63 sites.
- In the US, there were 44 sites (30 PDC, 14 non-PDC sites); of the 105 patients randomized in the US, 83 (79% of all US participants and 53% of all DINAMO participants) were randomized by PDC sites and 22 by non-PDC US sites.
- The main reason for screen failure was HbA1c out-of-range (57%); 25 participants were eligible for re-screening due to an initial modifiable exclusion (e.g., HbA1c <6.5%); 11 of whom were eventually randomized after re-screening.
- The study will be completed in fall 2022.

Figure 2. DINAMO: A global clinical trial

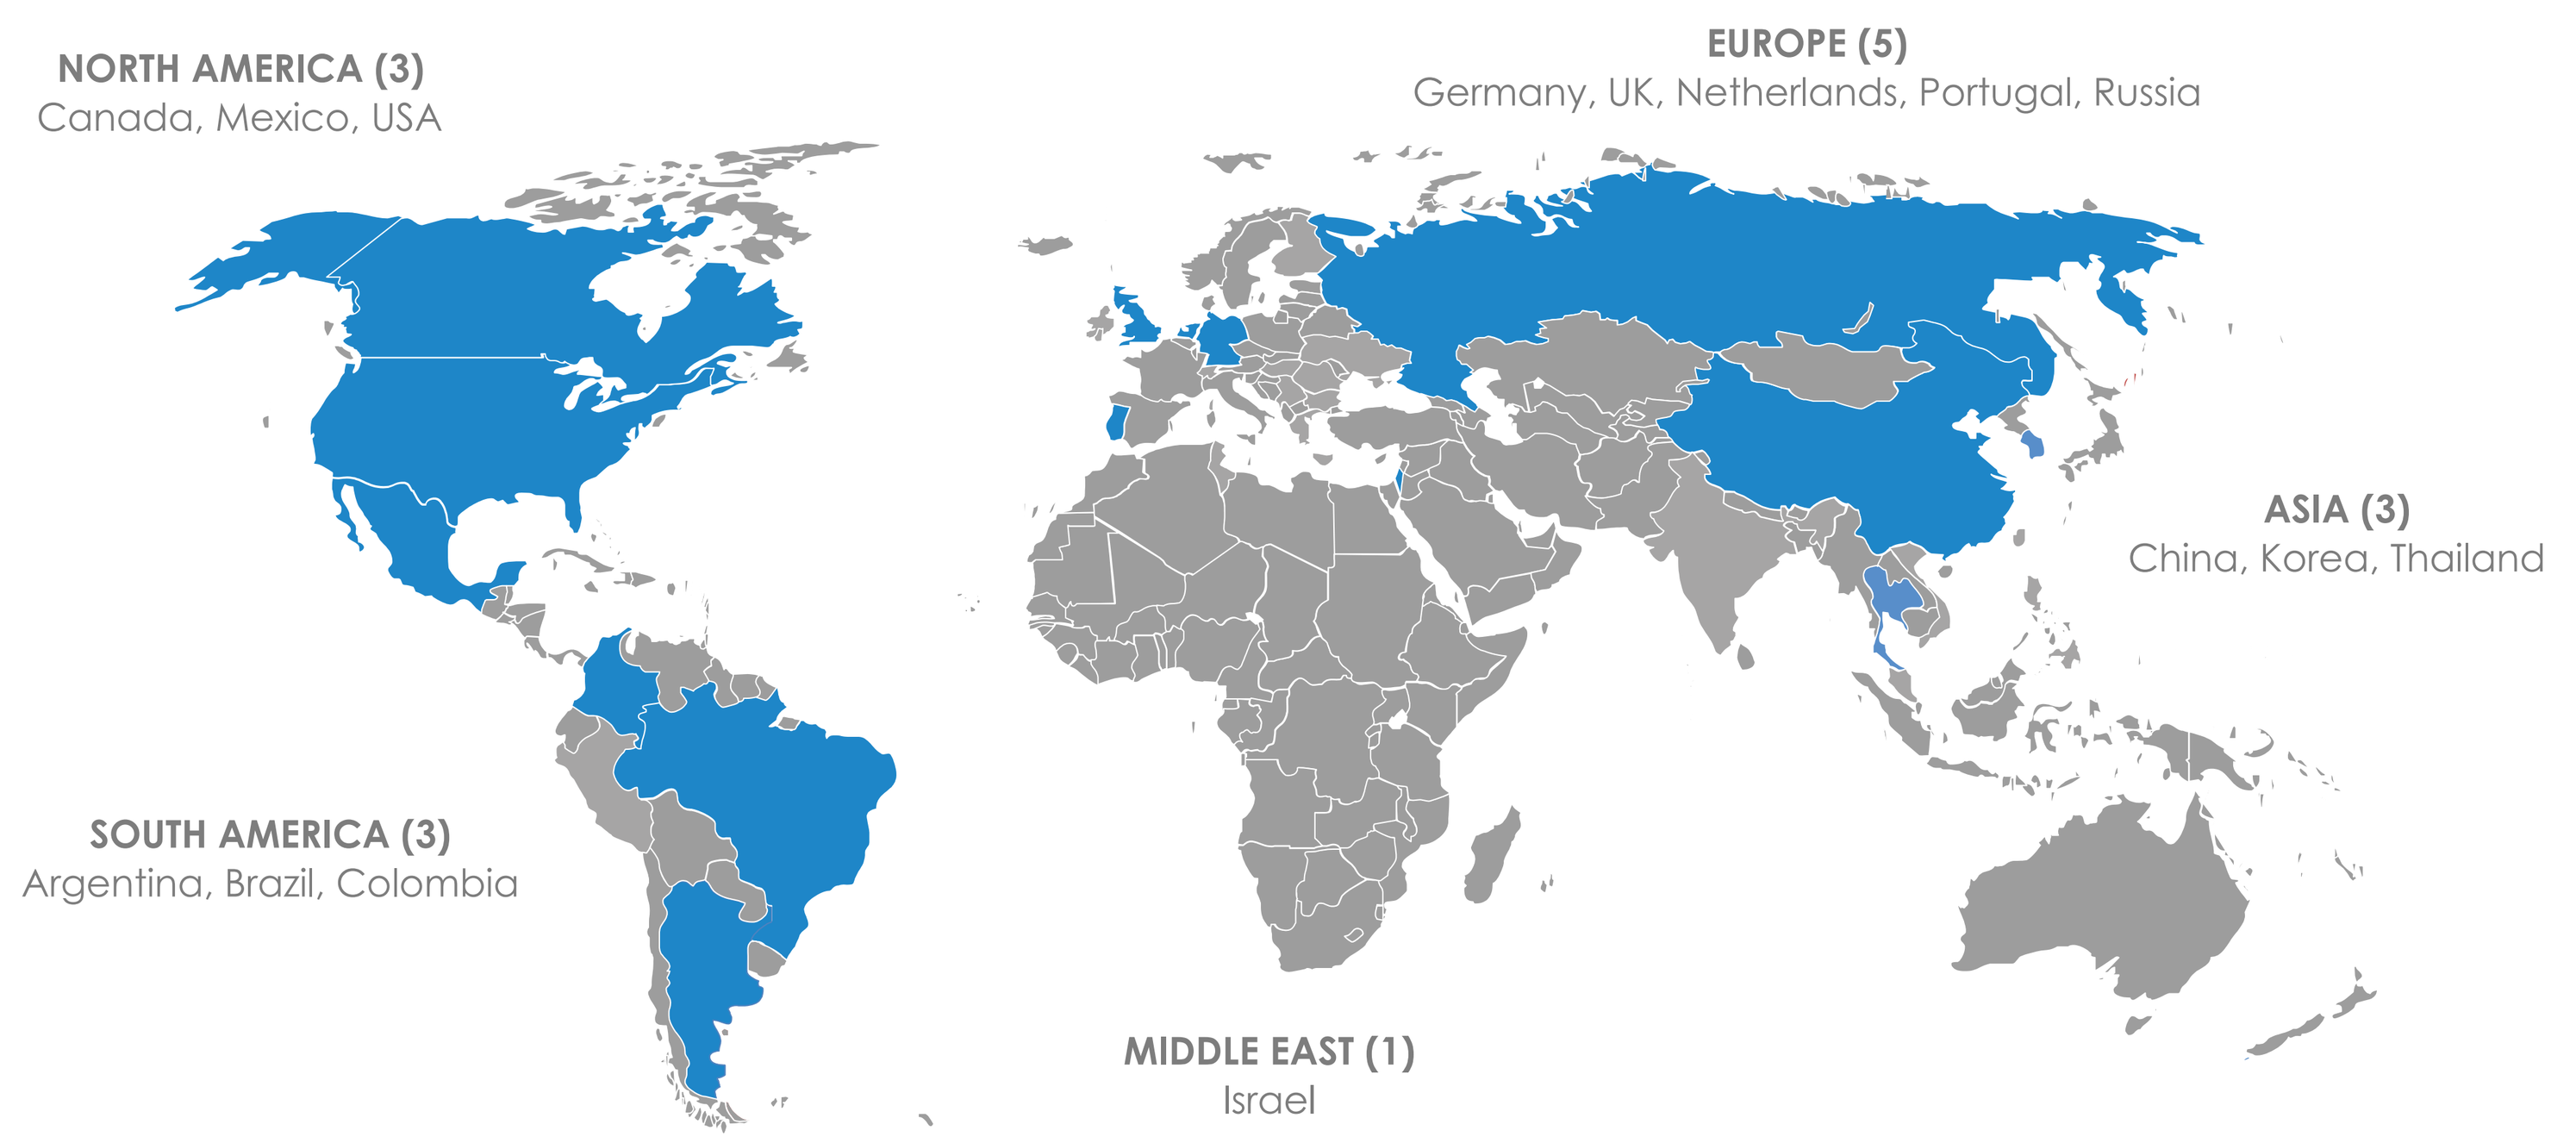
NORTH AMERICA (3)
Canada, Mexico, USA
EUROPE (5)
Germany, UK, Netherlands, Portugal, Russia
ASIA (3)
China, Korea, Thailand
SOUTH AMERICA (3)
Argentina, Brazil, Colombia
MIDDLE EAST (1)
Israel

Adaptations due to the COVID-19 pandemic

- Screening for the DINAMO trial was placed on hold from March to July 2020. Therefore, the participant enrollment timeline was extended from January to May 2021.
- During the COVID-19 pandemic, participants in screening who could adhere to onsite visit schedules and study medication dispensing were allowed to continue to randomization. If this was not feasible, the participant was considered a screen fail and allowed to rescreen when new participant enrollment resumed.
- Participants on treatment or in follow-up were required to complete primary/secondary endpoint visits onsite, if possible. For all other visits, if onsite visits were not possible, alternative processes were implemented to support the participant, such as phone visits, local laboratory testing, and delivery of study medication via courier.

Figure 3. DINAMO screening and randomization over time

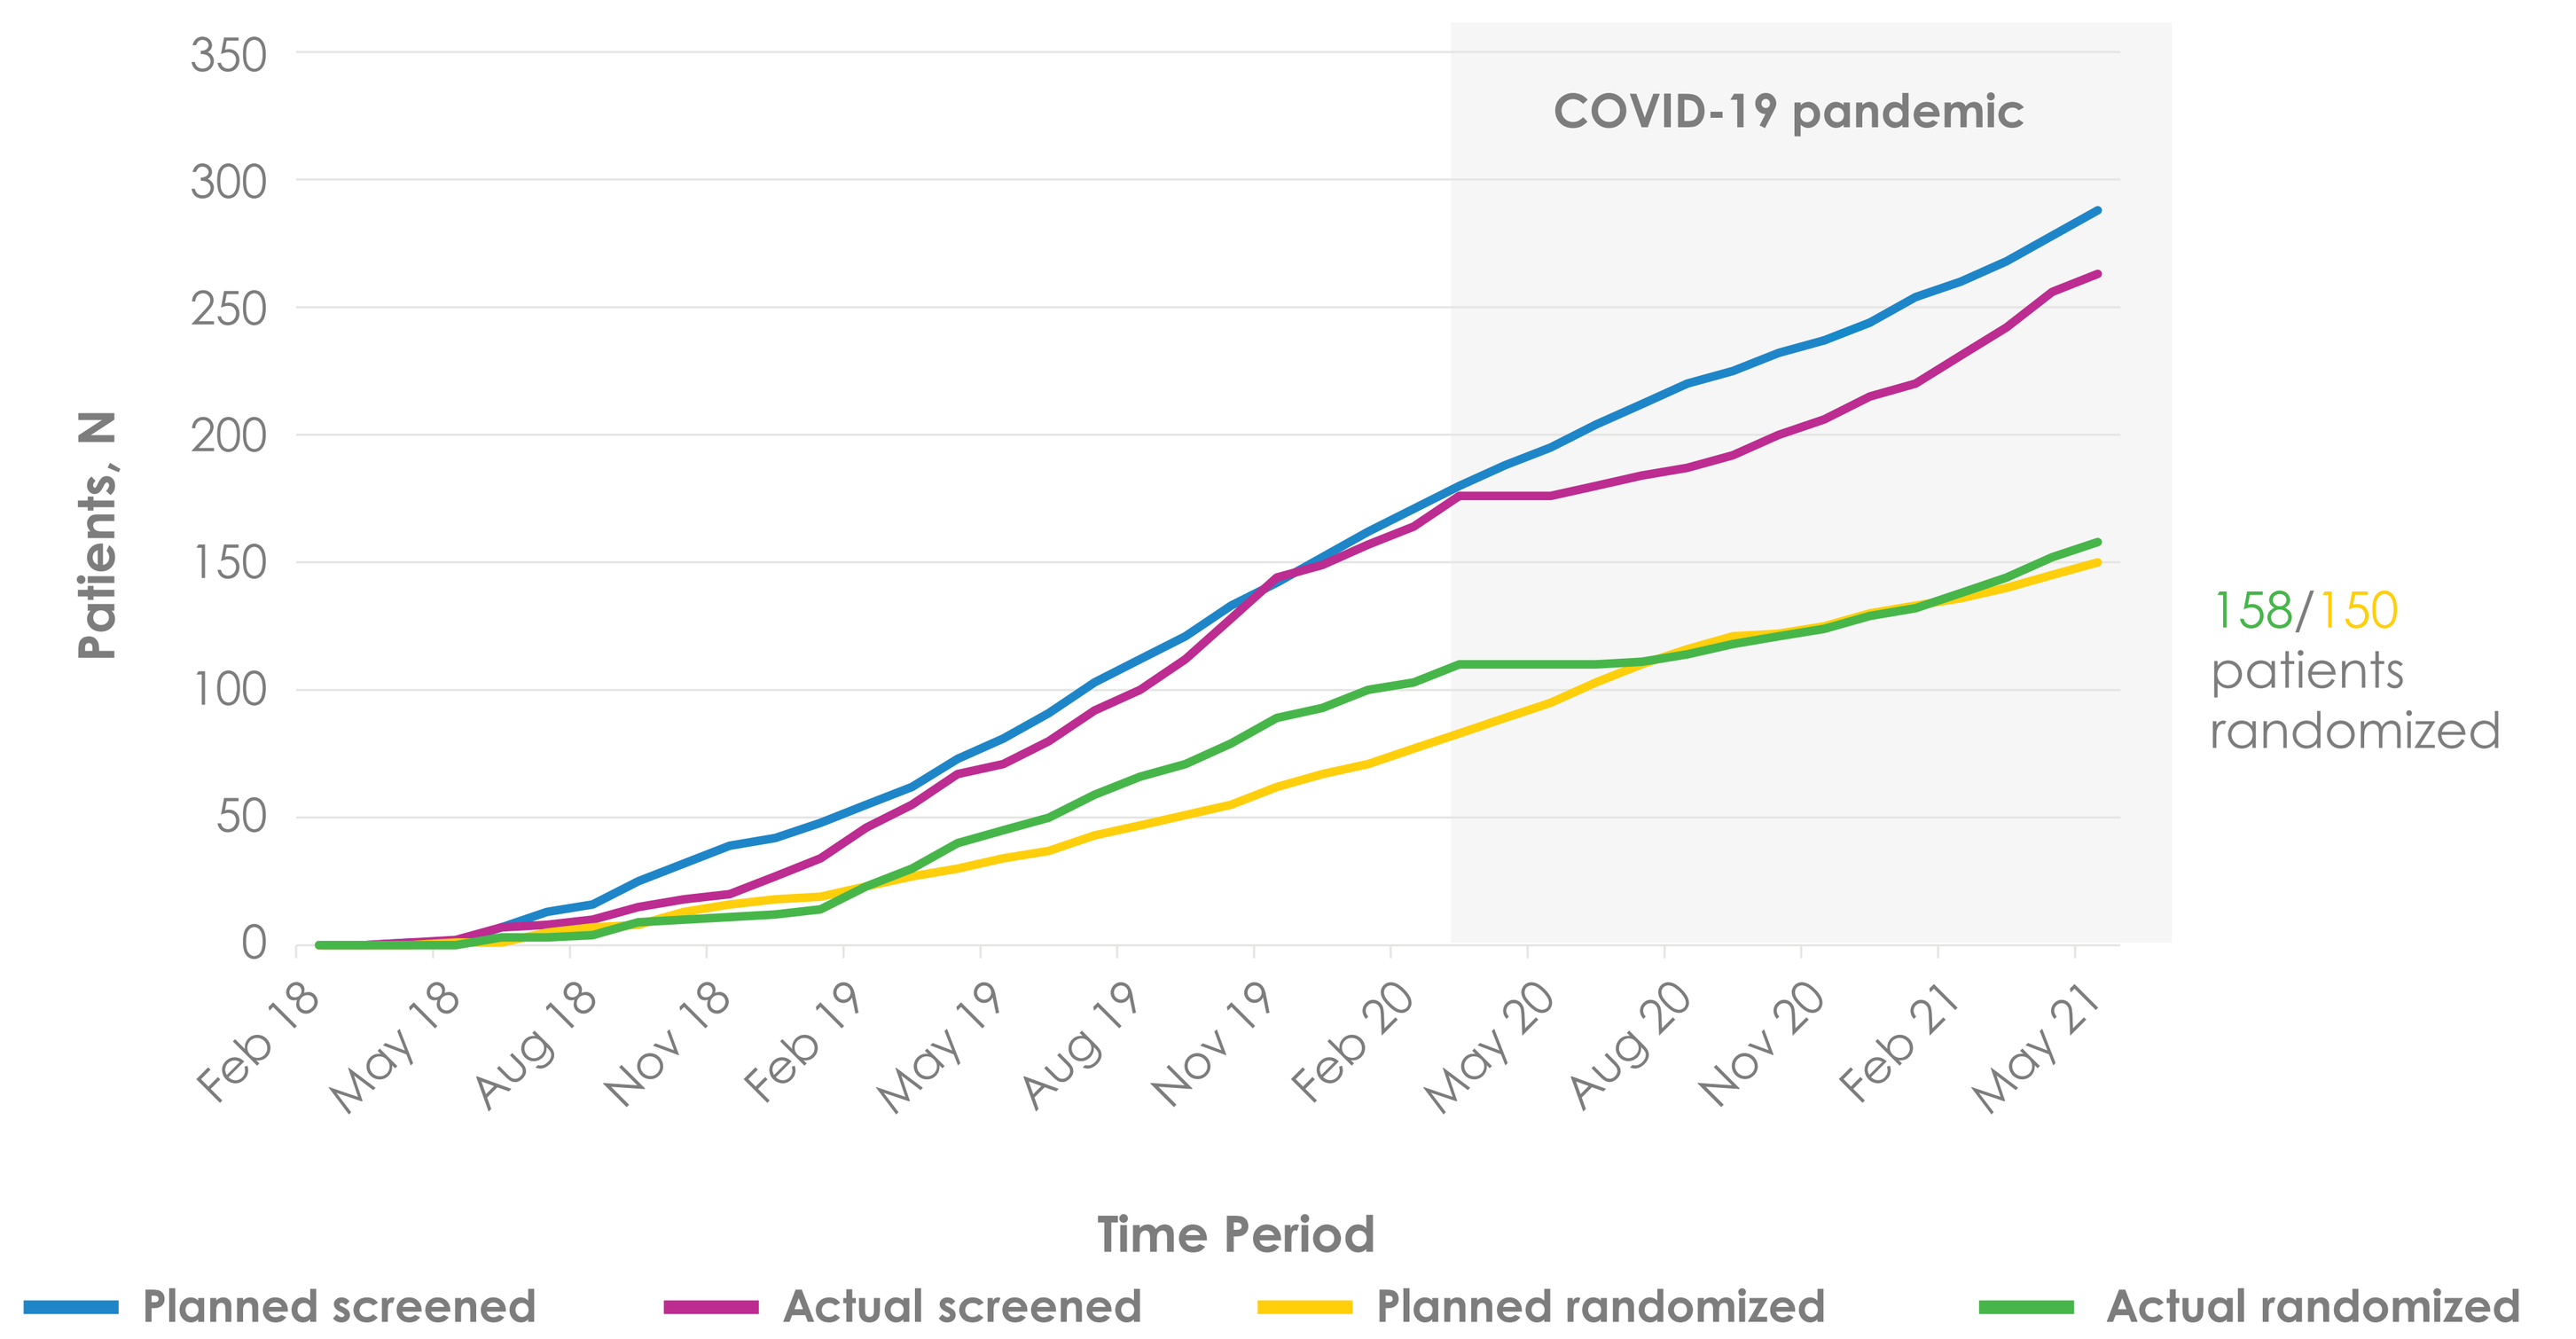
COVID-19 pandemic
Patients, N
350
300
250
200
150
100
50
0
Feb 18
May 18
Aug 18
Nov 18
Feb 19
May 19
Aug 19
Nov 19
Feb 20
May 20
Aug 20
Nov 20
Feb 21
May 21
Time Period
158/150
patients
randomized
Planned screened
Actual screened
Planned randomized
Actual randomized

Table 1. Screening data and main baseline characteristics

Screening data and baseline characteristics	DINAMO	US	Non US
Screened, N	263	156	107
Re-screened, N	25	13	12
Screen failure, N	105	51	54
Baseline characteristics (based on randomized set)			
Randomized, N (% pre/post COVID-19 pandemic*)	158 (70/30)	105 (69/31)	53 (74/26)
Age, years, mean (% <15 years old)	14.5 (49)	14.5 (50)	14.5 (47)
BMI, kg/m², mean ± SD	36.1 ± 8.3	38.0 ± 7.6	32.2 ± 8.4
BMI, SDS; Z-score, mean ± SD	2.97 ± 0.85	3.21 ± 0.68	2.50 ± 0.95
Female sex, %	61	62	60
Race, %			
American Indian or Alaska Native	5	3	9
Asian	6	1	15
Black or African-American	31	41	11
Native Hawaiian or other Pacific Islander	2	3	0
Multiple or missing information	6	5	9
White	50	48	55
Ethnicity, %			
Hispanic or Latino	39	34	47
Non-Hispanic	61	66	53
Region, %			
North America/South America/Europe/Asia	68/17/11/3	100/0/0/0	6/51/34/9
HbA1c, %, mean ± SD	8.0 ± 1.2	8.1 ± 1.2	7.9 ± 1.2
HbA1c, mmol/mol, mean ± SD	64 ± 13	65 ± 13	63 ± 13
Antidiabetes treatment, %			
Metformin/insulin/metformin and insulin/none	49/3/40/8	45/2/45/9	59/6/30/6

*Participants randomized up to March 17, 2020 were considered 'pre COVID', otherwise 'post COVID'.
BMI, body mass index; HbA1c, glycated hemoglobin; SD, standard deviation; SDS, standard deviation scores.

Comparisons of US with non-US participants

- Overall, baseline characteristics of DINAMO US participants versus non-US participants were generally comparable (**Table 1**).
- Of note, US participants had a higher BMI Z-score (3.21 vs 2.50), slightly higher HbA1c (8.1% vs 7.9%), and more insulin treatment at baseline (47% vs 36%).
- There was a higher proportion of non-White and non-Hispanic participants in US versus non-US participants.

CONCLUSIONS

- To our knowledge, this is the first pediatric T2D study to achieve patient recruitment consistent with the initially projected timeline.
- Innovative approaches to clinical research in children and adolescents with T2D, such as testing multiple active agents versus a single placebo group, re-screening of participants with modifiable exclusion criteria to mitigate high failure screening rates, a consortium model of multiple centers working together, and a dedicated Steering Committee involved in execution and monitoring of the study, can effectively overcome the many challenges of successfully completing pivotal trials.

Poster 994-P: 14-A Pediatrics—Obesity and Type 2 Diabetes, American Diabetes Association (ADA) 82nd Scientific Sessions 2022, June 3–7; New Orleans, LA, USA; Hybrid
Presenter: Dr Lori Laffel, Joslin Diabetes Center, Harvard Medical School, Boston, MA, USA; email: lori.laffel@joslin.harvard.edu

References

1. Tamborlane WV, *et al.* Liraglutide in Children and Adolescents with Type 2 Diabetes. *N Engl J Med.* 2019;381:637–646.
2. Tamborlane WV, *et al.* 1-LB: Once-Weekly Exanatide in Youth with Type 2 Diabetes: A Pivotal Phase III Randomized Study. *Diabetes* 1 June 2021 70 (Supplement_1).

SC-US-74433

Disclosures

LL: Advisor/Consultant for Janssen, Insulet, Boehringer Ingelheim (BI), Medtronic, Dompe, Provention, Eli Lilly (EL), Roche, Dexcom. **TD:** Speaker, advisory panel or research support from Astra Zeneca, Bayer, BI, Dexcom, EL, Insulet, Lifescan, Medtronic, Novo Nordisk (NN), Roche, Sanofi; shareholder from DreaMed Diabetes, Ltd. **WT:** Member of Steering Committee for DINAMO study. **GK:** No disclosures to report. **CS**, **DN**, **NS** and **JM** are employees of BI. **PZ:** Consulting for BI, Merck, EL, Janssen, I-ACT, NN. **SW:** Member of Steering Committee for DINAMO study. Served on a data safety monitoring board for the National Institute of Diabetes and Digestive and Kidney Diseases/NIH and serving on an advisory panel for Roche Diagnostics outside the submitted work. Served on an advisory board in health equity for Medtronic MiniMed.

Acknowledgments

The DINAMO trial was funded by Boehringer Ingelheim (BI). Editorial support, supported financially by BI, was provided by Elevate Scientific Solutions. The authors were fully responsible for all content and editorial decisions, were involved at all stages of poster development, and have approved the final version.