Spesolimab treatment improves pain, symptoms of psoriasis, fatigue and quality of life in patients with generalized pustular psoriasis: Patient-reported outcomes from the Effisayil™ 1 study

P1047

Alexander A. Navarini[1], Manuelle Viguier[2], Tsen-Fang Tsai[3], Akimichi Morita[4], Kristian Reich[5], Na Hu[6], Mogana Sivalingam[7], Christian Thoma[8], Mark G. Lebwohl[9]

[1]Department of Dermatology, University Hospital of Basel, Basel, Switzerland; [2]Department of Dermatology, Hôpital Robert Debré, Reims, France; [3]Department of Dermatology, National Taiwan University Hospital and National Taiwan University College of Medicine, Taipei, Taiwan; [4]Department of Geriatric and Environmental Dermatology, Nagoya City University, Graduate School of Medical Sciences, Nagoya, Japan; [5]Center of Translational Research in Inflammatory Skin Diseases, Institute for Health Services Research in Dermatology and Nursing, University Medical Center Hamburg-Eppendorf, Hamburg, Germany; [6]Boehringer Ingelheim Investment Co., Ltd, Shanghai, China; [7]Boehringer Ingelheim International GmbH, Ingelheim, Germany; [8]Boehringer Ingelheim International GmbH, Biberach, Germany; [9]Icahn School of Medicine at Mount Sinai, New York City, NY, USA

Patients with a generalized pustular psoriasis (GPP) flare who were treated with up to three doses of 900 mg of spesolimab intravenously showed clear improvements from baseline in the patient-reported outcomes (PROs) of pain, fatigue, overall quality of life (QoL) and skin symptoms

PURPOSE

To evaluate PROs of measures of pain, fatigue, impact on overall QoL and skin symptoms in patients treated with spesolimab, an anti-interleukin-36 receptor monoclonal antibody, in the Effisayil™ 1 study.

INTRODUCTION

- GPP is a rare, potentially life-threatening, neutrophilic skin disease characterised by widespread eruption of sterile, visible pustules, and can occur with or without systemic inflammation[1–3]
- In the multicentre, randomised, double-blind, placebo-controlled Effisayil™ 1 study (NCT03782792) in patients presenting with a GPP flare, spesolimab treatment led to rapid pustular and skin clearance within 1 week.[4] GPP flares are associated with a high clinical burden in PROs including symptoms such as pain, itching and fatigue, which all impact overall QoL[5,6]
- Here we explore PROs in patients with a GPP flare receiving spesolimab treatment

CONCLUSIONS

- In this study, patients who received 900 mg of intravenous spesolimab showed clinically significant improvements from baseline in the PROs of pain, fatigue, overall QoL and skin symptoms
- The clear separation of the spesolimab and placebo curve occurred early during the placebo-controlled period (Week 1). This suggests that spesolimab results in the rapid improvement of PROs, with considerable improvement in fatigue and pain
- PRO scores continued to improve up to Week 4 and were sustained through Week 12

METHODS

- Patients (N=53) were randomised (2:1) to receive placebo or 900 mg of spesolimab on Day 1 and were followed for 12 weeks
- If disease worsening occurred during Week 1, patients were able to receive escape treatment (any other treatment for GPP) any time after their first dose of spesolimab or placebo on Day 1 and before Day 8
- Patients who achieved qualifying clinical assessment scores (GPPGA total score ≥2 or GPPGA pustulation subscore ≥2) and did not receive escape treatment during Week 1 were eligible to receive open-label (OL) spesolimab on Day 8 and another dose of OL spesolimab between Day 8 and Week 12 to treat new flares
 - Spesolimab arm (n=35): One dose of spesolimab at Day 1 (n=23); optional second dose of OL spesolimab at Day 8 (n=12); optional third dose of OL spesolimab between Weeks 1 and 12 (n=2)
 - Placebo arm (n=18): optional first dose of OL spesolimab at Day 8 (n=15); optional second dose of OL spesolimab between Weeks 1 and 12 (n=1)
- All randomised patients were included in this analysis. The observed cases irrespective of any use of escape treatment, OL spesolimab on Day 8 or after Day 8 (representing the intention-to-treat principle) are summarised descriptively
- To monitor any changes in outcomes, patients completed the following PRO questionnaires throughout the study: Pain visual analogue scale (pain VAS), Functional Assessment of Chronic Illness Therapy–Fatigue (FACIT-Fatigue), Dermatology Life Quality Index (DLQI) and the Psoriasis Symptom Scale (PSS)
 - All four PROs were measured on Day 1, Day 8 and Weeks 2–4, 8 and 12. PSS scores were also measured on Day 2 and Day 3

RESULTS

PRO survey multiple-choice answers and their corresponding scores

Pain VAS
- Measures severity of pain
- Score is determined by measuring the distance (mm) on the 10 cm line between the 'no pain' anchor and the patient's mark

No pain — Very severe pain
0 — 10 cm — 100

DLQI
- 10-item scale
- Assesses six areas of quality of life: symptoms and feelings, daily activities, leisure, work and school, personal relationships and treatment
- Scores from each answer totalled; maximum 30

No effect (0–1) — Small effect (2–5) — Moderate effect (6–10) — Very large effect (11–20) — Extremely large effect (21–30)
0 — 30

FACIT-Fatigue
- 13-item scale
- Measures self-reported fatigue and its impact on daily activities and function
- Scores from each answer totalled; maximum 52

High impact — Low impact
0 — 52

PSS
- 4-item scale
- Measures severity of pain, redness, itching and burning
- Scores from each answer totalled; maximum 16

Less severe — More severe
0 — 16

DLQI, Dermatology Life Quality Index; FACIT-Fatigue, Functional Assessment of Chronic Illness Therapy–Fatigue; PRO, patient-reported outcome; PSS, Psoriasis Symptom Scale; VAS, visual analogue scale.

High total scores indicate a large impairment or intense severity, except for FACIT-Fatigue, for which higher scores represent less fatigue

Absolute change from baseline in PRO scores over time

Pain VAS

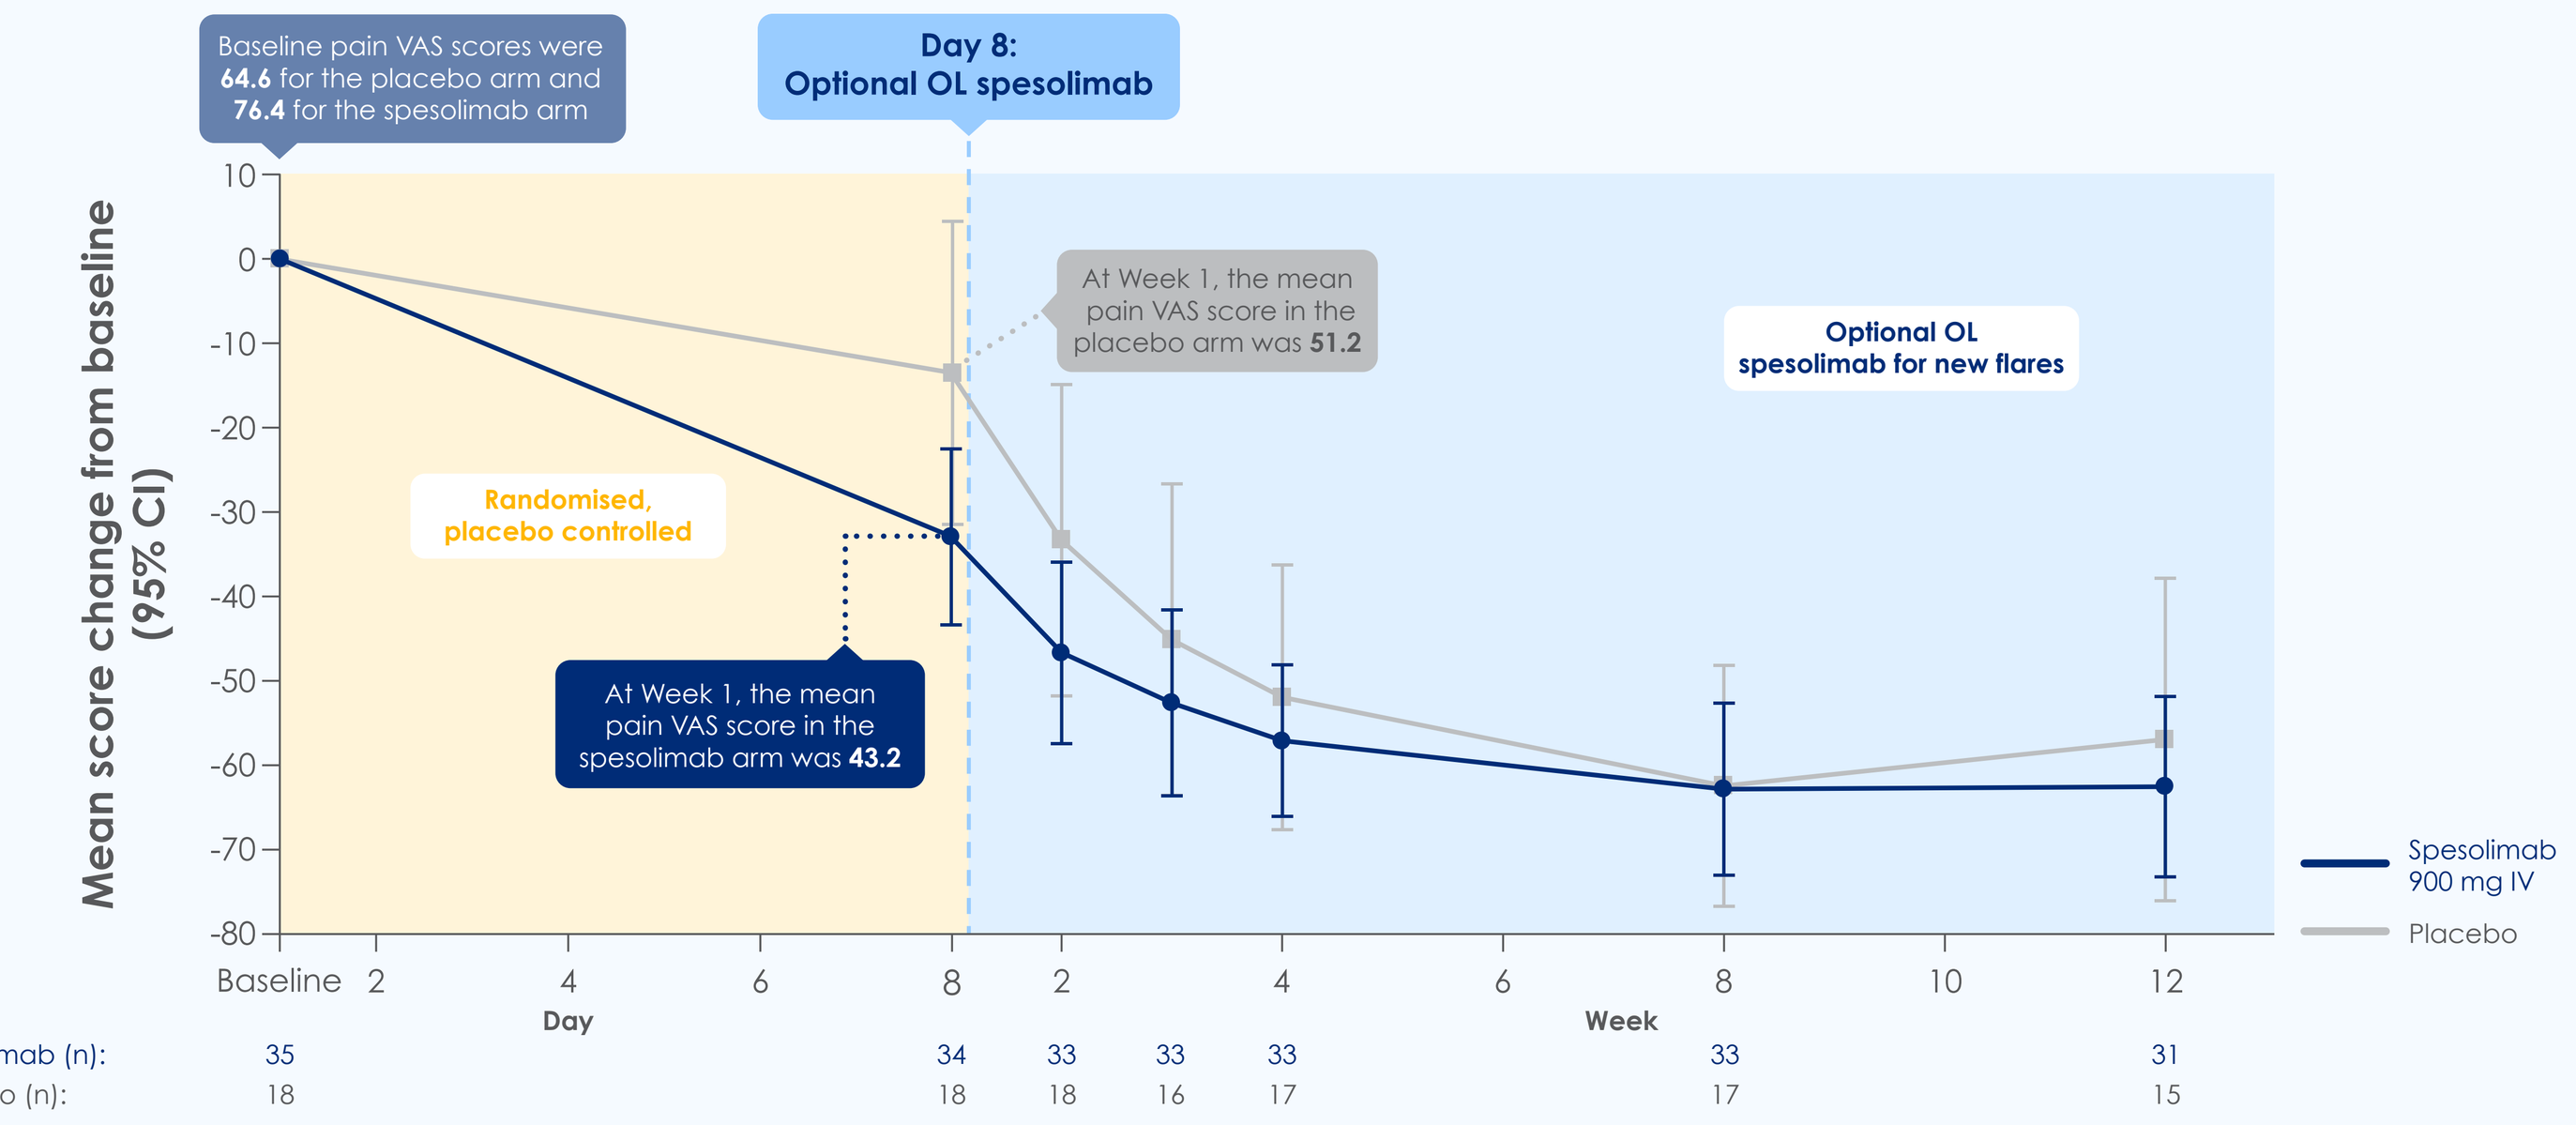

Baseline pain VAS scores were 64.6 for the placebo arm and 76.4 for the spesolimab arm
Day 8: Optional OL spesolimab
Mean score change from baseline (95% CI)
Randomised, placebo controlled
At Week 1, the mean pain VAS score in the placebo arm was 51.2
At Week 1, the mean pain VAS score in the spesolimab arm was 43.2
Optional OL spesolimab for new flares
Spesolimab 900 mg IV
Placebo
Day
Week
Spesolimab (n): 35 34 33 33 33 33 31
Placebo (n): 18 18 18 16 17 17 15

FACIT-Fatigue

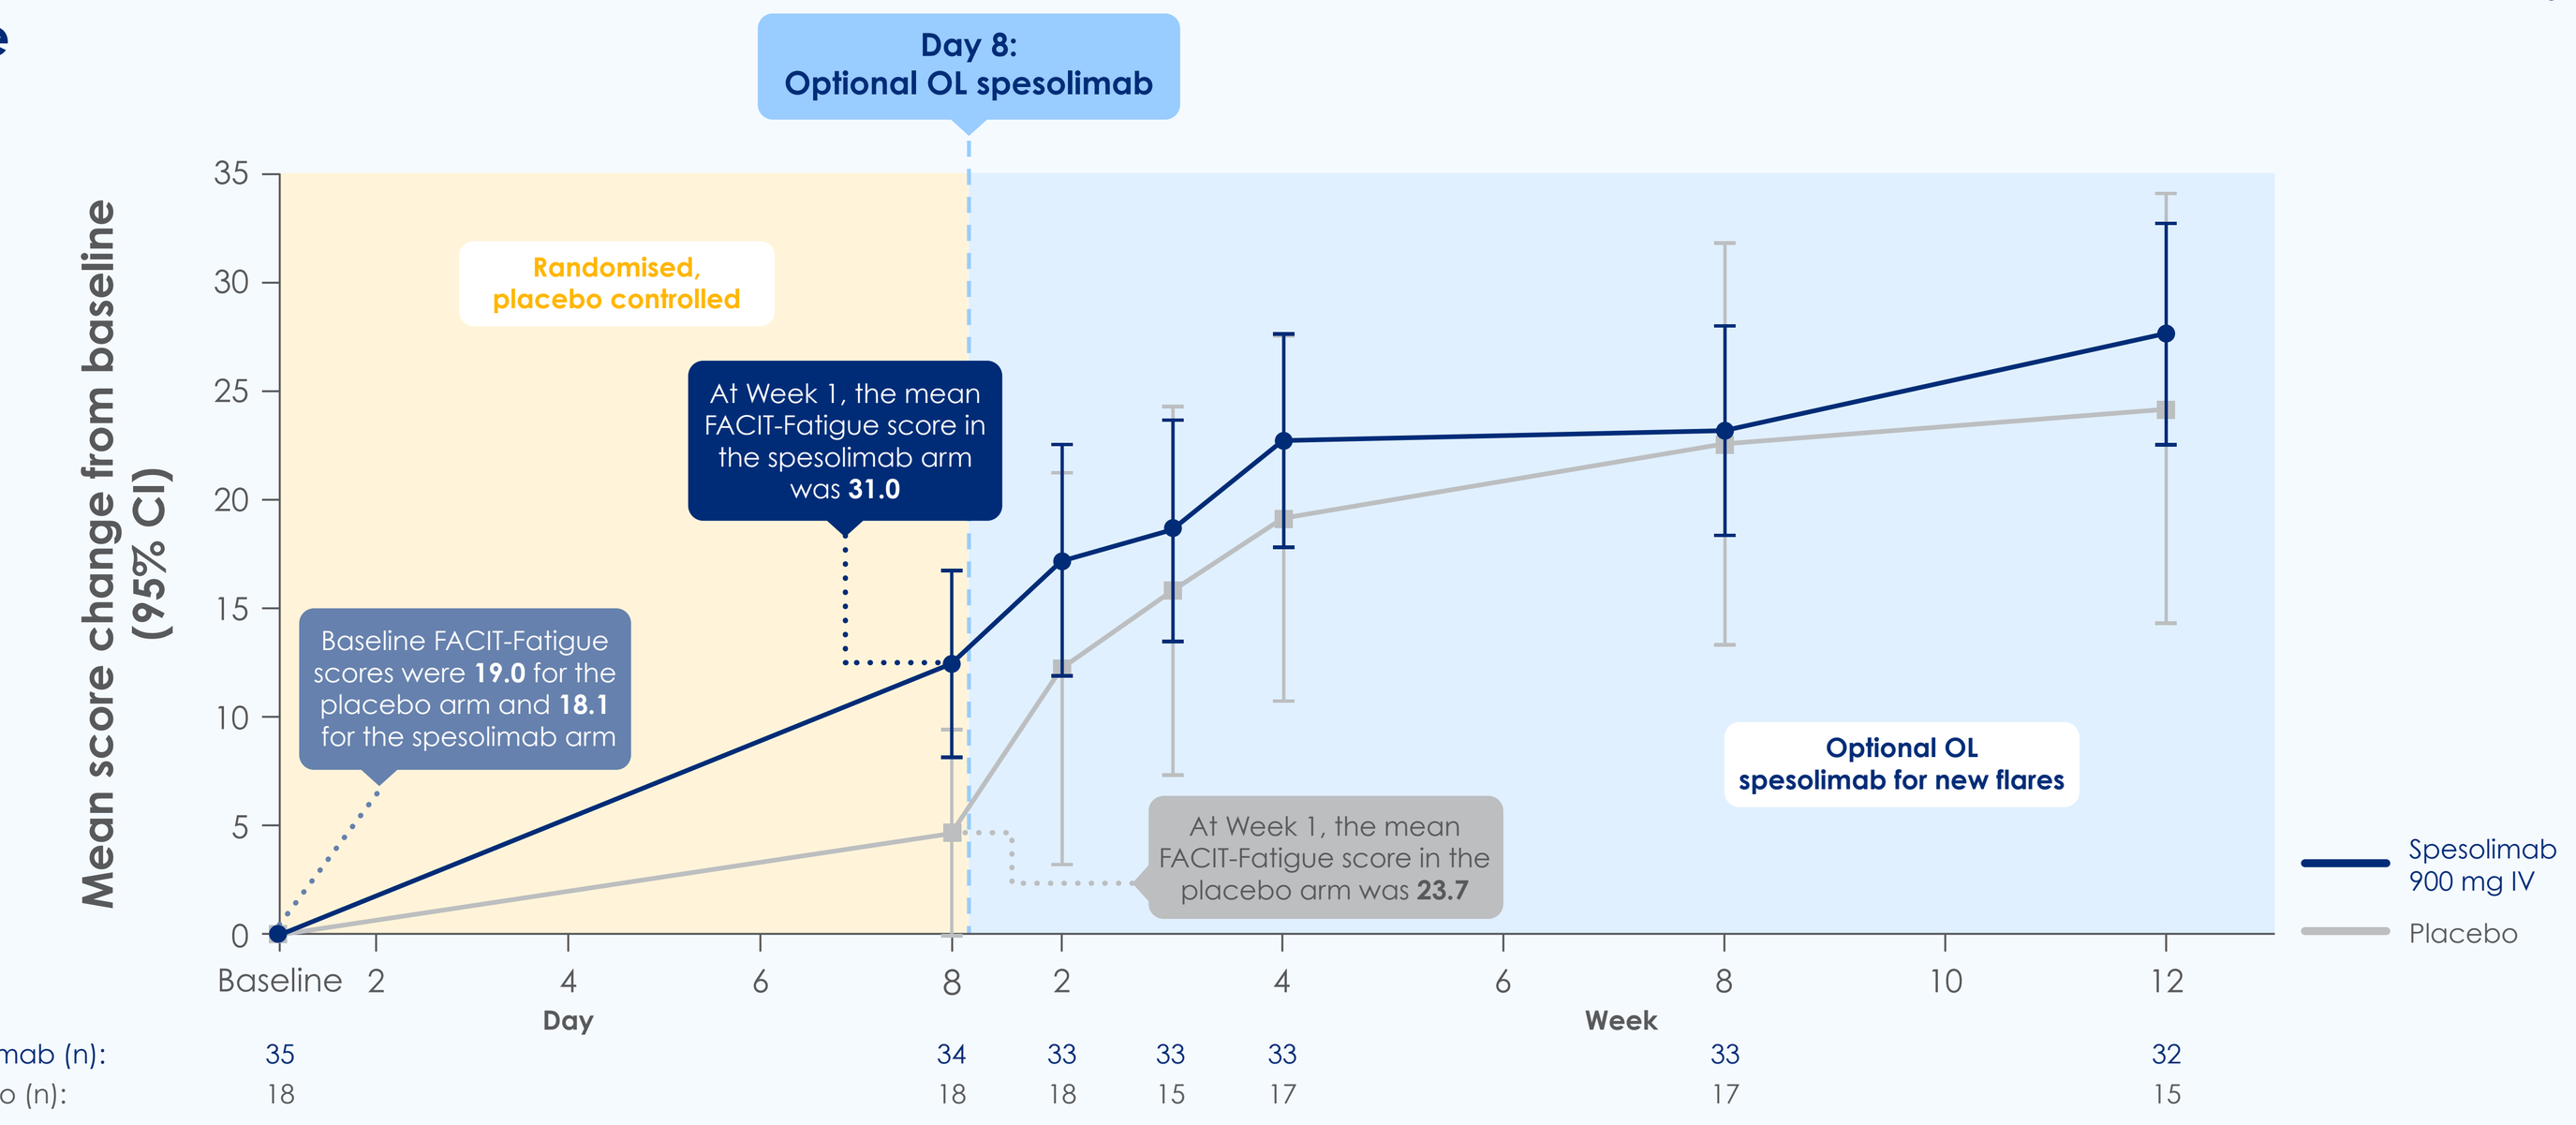

Day 8: Optional OL spesolimab
Mean score change from baseline (95% CI)
Randomised, placebo controlled
At Week 1, the mean FACIT-Fatigue score in the spesolimab arm was 31.0
Baseline FACIT-Fatigue scores were 19.0 for the placebo arm and 18.1 for the spesolimab arm
At Week 1, the mean FACIT-Fatigue score in the placebo arm was 23.7
Optional OL spesolimab for new flares
Spesolimab 900 mg IV
Placebo
Day
Week
Spesolimab (n): 35 34 33 33 33 33 32
Placebo (n): 18 18 18 15 17 17 15

DLQI

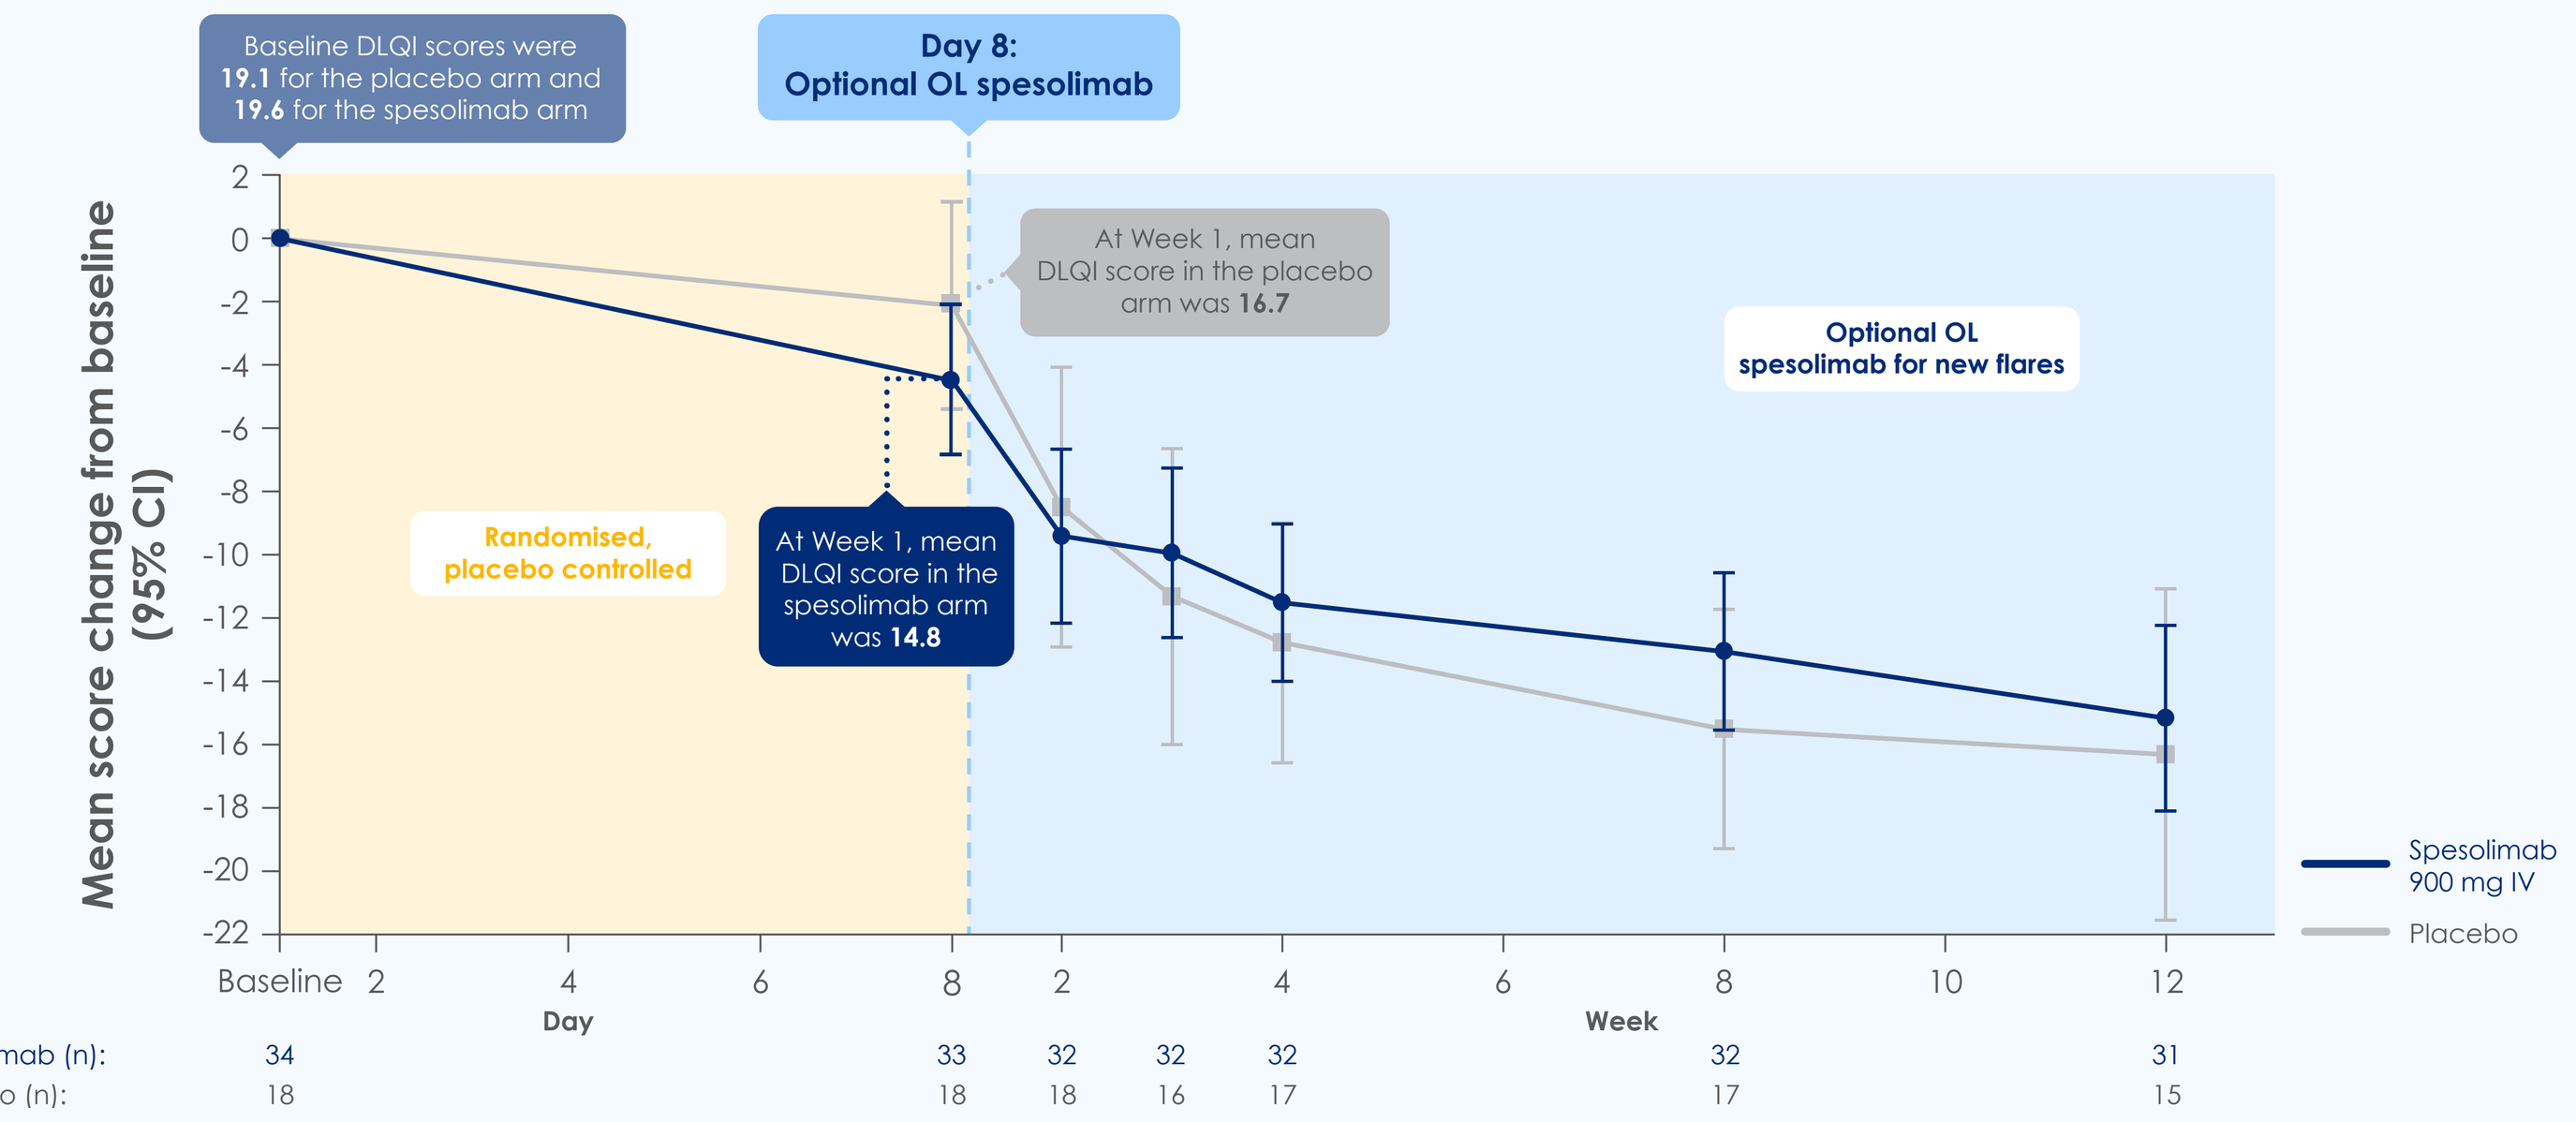

Baseline DLQI scores were 19.1 for the placebo arm and 19.6 for the spesolimab arm
Day 8: Optional OL spesolimab
Mean score change from baseline (95% CI)
At Week 1, mean DLQI score in the placebo arm was 16.7
Randomised, placebo controlled
At Week 1, mean DLQI score in the spesolimab arm was 14.8
Optional OL spesolimab for new flares
Spesolimab 900 mg IV
Placebo
Day
Week
Spesolimab (n): 34 33 32 32 32 32 31
Placebo (n): 18 18 18 16 17 17 15

PSS

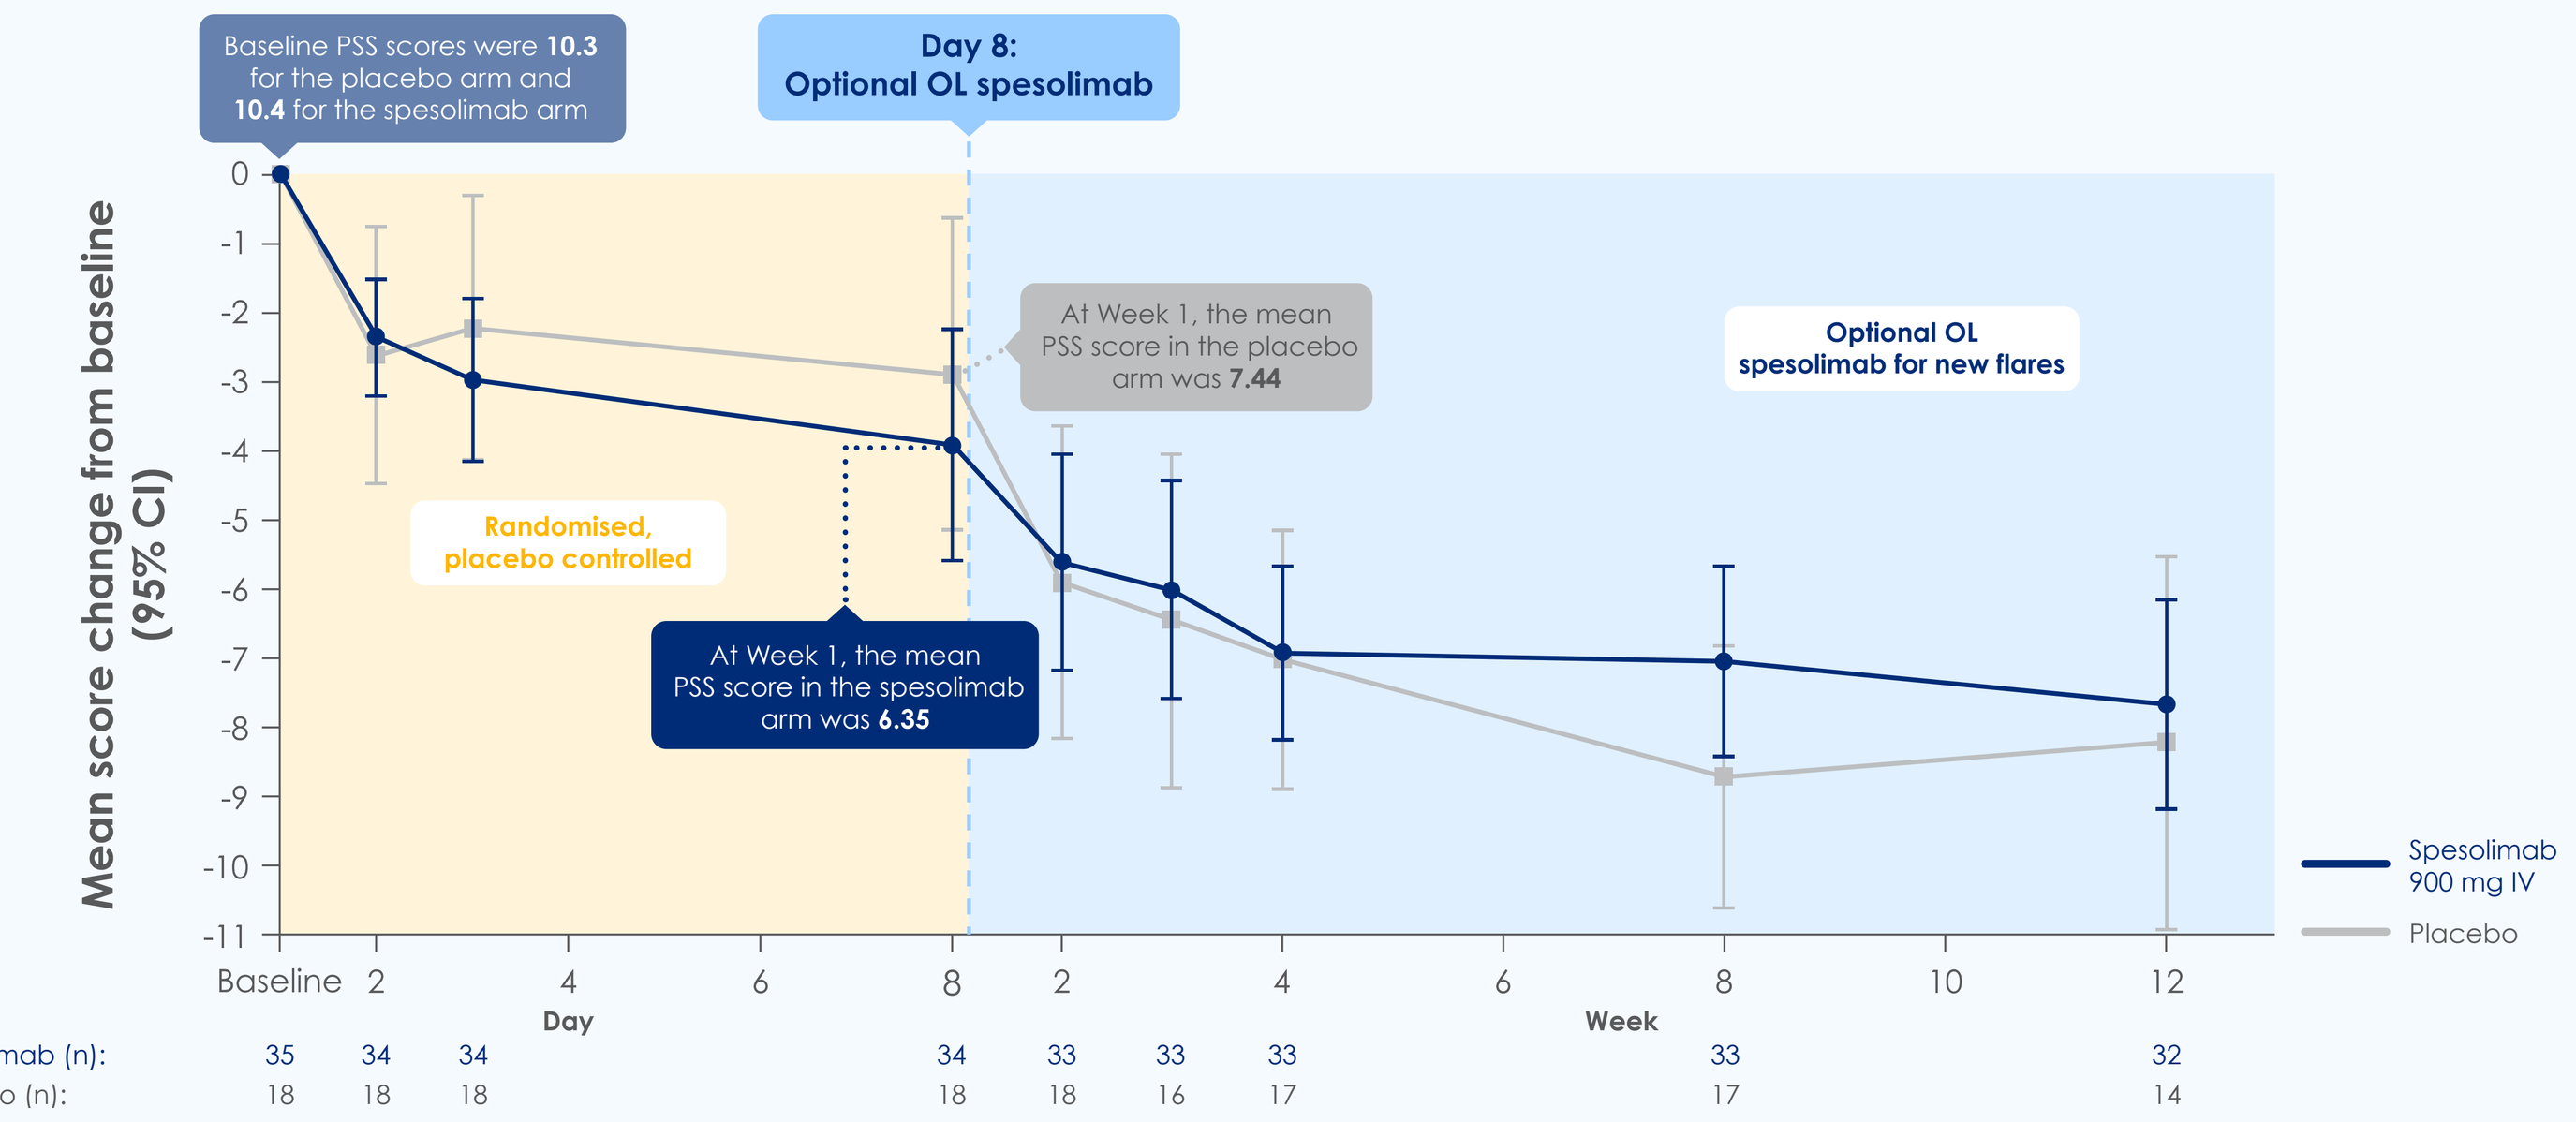

Baseline PSS scores were 10.3 for the placebo arm and 10.4 for the spesolimab arm
Day 8: Optional OL spesolimab
Mean score change from baseline (95% CI)
At Week 1, the mean PSS score in the placebo arm was 7.44
Randomised, placebo controlled
At Week 1, the mean PSS score in the spesolimab arm was 6.35
Optional OL spesolimab for new flares
Spesolimab 900 mg IV
Placebo
Day
Week
Spesolimab (n): 35 34 34 34 33 33 33 33 32
Placebo (n): 18 18 18 18 18 16 17 17 14

CI, confidence interval; DLQI, Dermatology Life Quality Index; FACIT-Fatigue, Functional Assessment of Chronic Illness Therapy–Fatigue; IV, intravenous; OL, open-label; PRO, patient-reported outcome; PSS, Psoriasis Symptom Scale; VAS, visual analogue scale.

Although the study was not designed to test a statistical difference for PROs between spesolimab and placebo earlier than week 4, there was a numerical trend of early separation between spesolimab and placebo within the first week which continued to improve over time with spesolimab. The placebo curve begins to converge with the spesolimab curve after administration of OL spesolimab at Day 8

References

1. Navarini AA, et al. *J Eur Acad Dermatol Venereol* 2017;31:1792–1799.
2. Bachelez H. *Acta Derm Venereol* 2020;100:adv00034.
3. Ryan TJ and Baker H. *Br J Dermatol* 1971;85:407–411.
4. Bachelez H, et al. WPPAC 2021;35129.
5. Choon SE, et al. *Int J Dermatol* 2014;53:676–684.
6. Strober B, et al. *J Am Acad Dermatol* 2021;85(Suppl.):AB108 (Abstract 26588).

Disclosures & Acknowledgements

This study was supported and funded by Boehringer Ingelheim. The authors met the criteria for authorship as recommended by the International Committee of Medical Journal Editors (ICMJE). The authors did not receive payment related to the development of the poster. Boehringer Ingelheim was given the opportunity to review the poster for medical and scientific accuracy as well as intellectual property considerations. AAN declares being a consultant and/or advisor and/or receiving speaker fees/grants and/or serving as an investigator in clinical trials for AbbVie, Almirall, Amgen, Bristol Myers Squibb (BMS), Boehringer Ingelheim, Celgene, Eli Lilly, Galderma, GlaxoSmithKline (GSK), LEO Pharma, Janssen-Cilag, Merck Sharp & Dohme (MSD), Novartis, Pfizer, Sandoz, Sanofi, Serono and UCB Pharma. MV declares receiving grants and consulting fees from Boehringer Ingelheim. T-FT declares conducting clinical trials and receiving honoraria for serving as a consultant for AbbVie, Boehringer Ingelheim, BMS, Celgene, Eli Lilly, Galderma, GSK, Janssen-Cilag, LEO Pharma, MSD, Novartis International, Pfizer and UCB Pharma. AM declares receiving research grants, consulting fees and/or speaker fees from AbbVie, Boehringer Ingelheim, Celgene, Eisai, Eli Lilly, Janssen, Kyowa Kirin, LEO Pharma, Maruho, Mitsubishi Tanabe, Nichi-Iko, Nippon Kayaku, Novartis, Sun Pharmaceutical Industries, Taiho Pharmaceutical, Torii Pharmaceutical and Ushio. KR has served as an advisor, speaker and investigator for AbbVie, Affibody, Almirall, Amgen, Avillion, Biogen, BMS, Boehringer Ingelheim, Celgene, Covagen, Eli Lilly, Forward Pharma, Fresenius Medical Care, Galapagos, GSK, Janssen-Cilag, Kyowa Kirin, LEO Pharma, Medac, MSD, Miltenyi Biotec, Novartis, Ocean Pharma, Pfizer, Regeneron, Samsung Bioepis, Sanofi, Sun Pharmaceutical Industries, Takeda, UCB Pharma, Valeant and XenoPort. NH, MS and CT are employees of Boehringer Ingelheim. MGL is an employee of Mount Sinai and declares receiving research funds from AbbVie, Amgen, Arcutis, Avotres, Boehringer Ingelheim, Dermavant Sciences, Eli Lilly, Incyte, Janssen Research & Development, Ortho Dermatologics, Regeneron and UCB Pharma, and paid consulting activities for Aditum Bio, Almirall, AltruBio, AnaptysBio, Arcutis, Aristea Therapeutics, Arrive Technologies, Avotres Therapeutics, BiomX, Boehringer Ingelheim, BMS, Cara Therapeutics, Castle Biosciences, Corrona, Dermavant Sciences, Dr. Reddy's Laboratories, Evelo Biosciences, Evommune, Facilitatation of International Dermatology Education, Forte Biosciences, Foundation for Research and Education in Dermatology, Helsinn Therapeutics, Hexima, LEO Pharma, Meiji Seika Pharma, Mindera, Pfizer, Seanergy and Verrica. Geetha Vilventhraraja, BSc, of OPEN Health Communications (London, UK), provided writing, editorial support and formatting assistance, which was contracted and funded by Boehringer Ingelheim.

Scan QR code for an interactive, electronic, device-friendly copy of this poster
https://bit.ly/3kjSdxl

Click the icon to access an interactive microsite for this Smart poster

30th European Academy of Dermatology and Venereology Congress (29 September – 2 October 2021, Virtual)